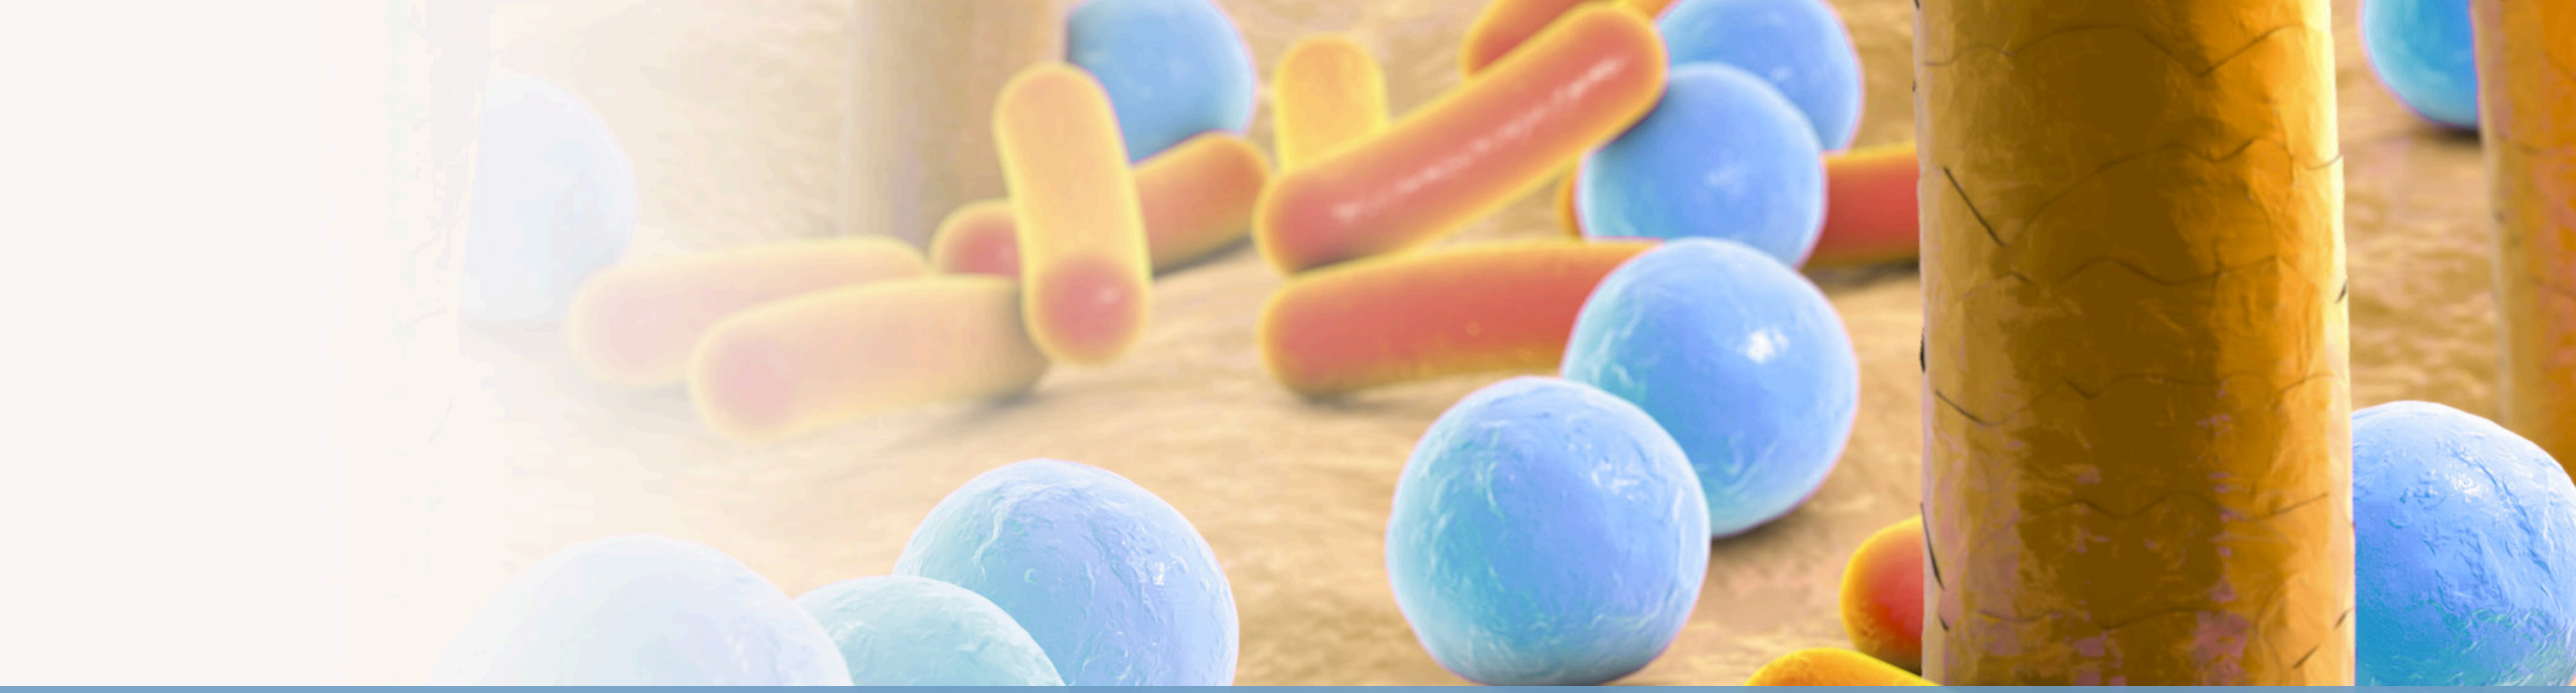

Effects of Topical Lotions on the Atopic Dermatitis Skin Microbiome and Associations with Itch and Skin Barrier Function

Kimberly Capone, Ph.D., Shifra Liba Klein, Ph.D., Frank Kirchner, M.S., Neena Tierney, Ph.D. Johnson & Johnson Consumer, Inc.

Presented at the 76th Annual Society for Investigative Dermatology (SID) Meeting, Portland, OR, April 26-29, 2017

PURPOSE

Atopic Dermatitis (AD) lesions are often colonized by *Staphylococcus aureus*, and improvements in microbial diversity are reported to correlate with improvements in disease severity. In addition, follicular colonization by *S. aureus* has been implicated in exacerbating the sensations of itch in the AD cycle. Despite these advances, the function of the skin microbiota in the pathogenesis of AD remains poorly understood, particularly in different patient populations. Advances in our understanding of the AD-associated skin microbiome along the disease progression will facilitate deeper insights into the function of the skin microbiota in this complex ecosystem, and ultimately novel microbiome-based therapies.

Therefore, we undertook to examine the changes in the skin microbiota, AD disease severity, and itch, during 14 days of treatment with two different topical lotions, including AVEENO® Eczema Therapy Moisturizing Cream (specially formulated for use on AD patients) and a non-fragranced lotion. The 14-day treatment phase was followed by a 7-day regression phase to observe alterations in the skin microbiota with worsening disease.

CLINICAL DESIGN

This multi-center, randomized, controlled 14-day clinical usage study was conducted to associate the skin microbiota of AD patients with pruritus, or itch, during the use of 2 different topical lotions. The patient population consisted of female and male subjects aged 16 – 50 years with mild to moderate atopic dermatitis, and a recent itch flare-up.

A total of 65 subjects participated in the study; 31 subjects used AVEENO® Eczema Therapy Moisturizing Cream (22 female, 9 male) and 34 subjects used the non-fragranced lotion (27 female, 7 male). Overall, 75% of the subjects were female and 25% male, with an average age of 34.5 years.

During the course of the study, subjects applied the assigned test material on all areas of the body (including face), as directed. Subjects were also provided with AVEENO® Baby Cleansing Therapy Moisturizing Wash to use in place of normal body wash and/or soap. Subjects used the test materials twice per day for 14 days followed by a 7-day regression period during which treatment was removed.

TABLE 1. Summary of Demographic Information (ITT Population)

	Total Subjects (N)		AVEENO® Eczema Therapy Moisturizing Cream		Non-fragranced Lotion	
N	65		31		34	
Mean Age (years)	34.5		34.8		34.2	
	N	(%)	N	(%)	N	(%)
Female	49	(75.4)	22	(71.0)	27	(79.4)
Male	16	(24.6)	9	(29.0)	7	(20.6)

Clinical evaluations were conducted at Baseline, Day 1, 3, 7, 14, and 21. Subjects underwent the following procedures at each of the indicated time points: clinical assessment including Eczema Area Severity Index (EASI) and Atopic Dermatitis Severity Index (ADSI), self-assessment questionnaires, including itch severity (Visual Analog Scale (VAS), digital imaging, non-invasive measures of skin (barrier function TEWL, skin hydration, pH), and microbiome sampling.

RESULTS

FIGURE 1. Skin Barrier Measurements

- Use of AVEENO® Eczema Therapy Moisturizing Cream resulted in a statistically significant decrease in pH from Baseline to Day 14 on lesional and non-lesional skin. Removal of treatment resulted in the pH rebounding. Limited skin pH improvements during treatment phase were seen for the non-fragranced lotion.

Skin pH for Lesion and Non-Lesion site

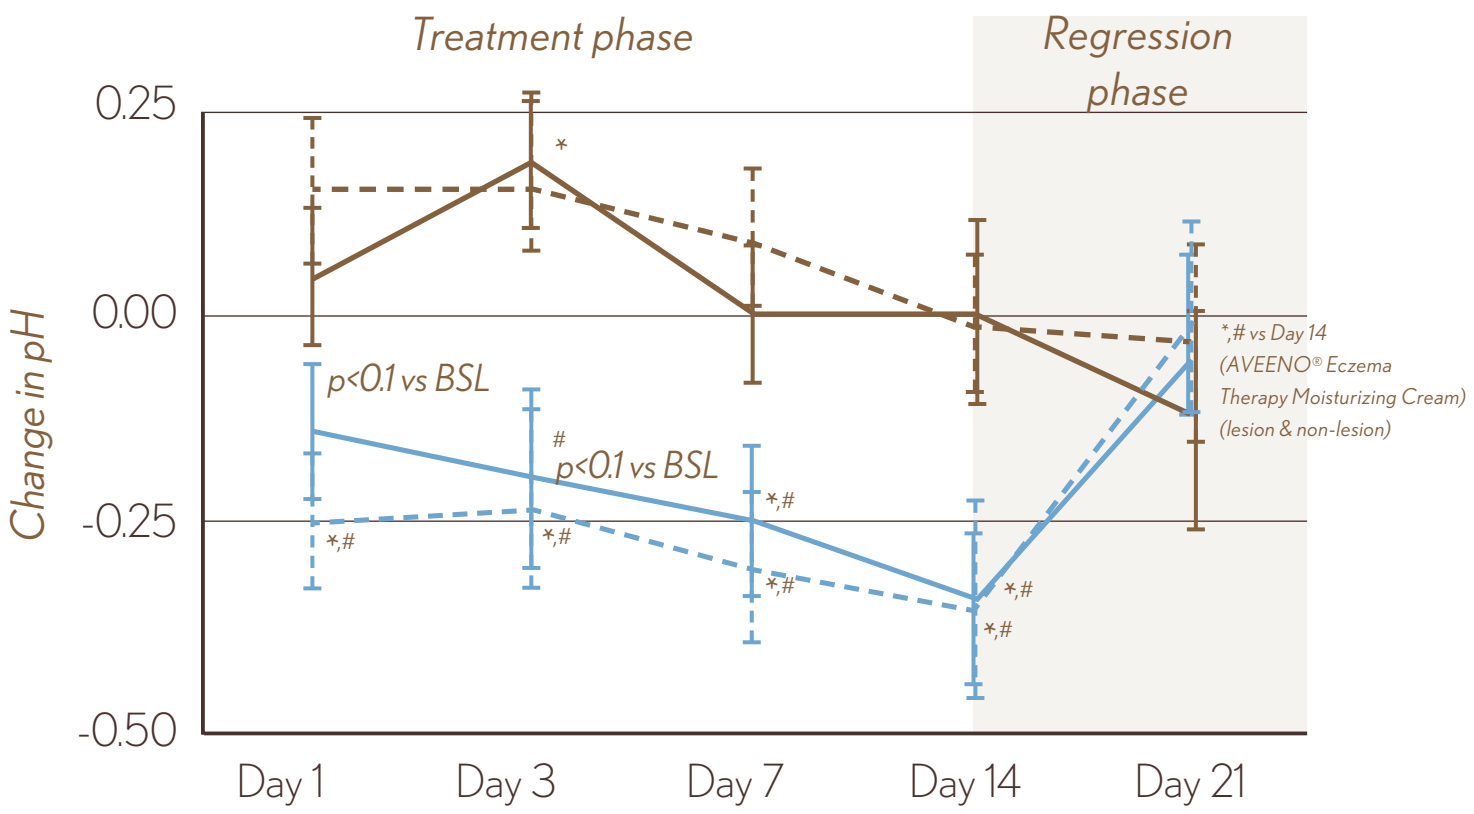
Treatment phase
Regression phase
Change in pH
0.25
0.00
-0.25
-0.50
p<0.1 vs BSL
#
p<0.1 vs BSL
*,#
*
*,# vs Day 14 (AVEENO® Eczema Therapy Moisturizing Cream) (lesion & non-lesion)
Day 1
Day 3
Day 7
Day 14
Day 21

- Use of AVEENO® Eczema Therapy Moisturizing Cream resulted in a statistically significant improvement in barrier function, as measured by a decrease in TEWL values for lesional skin at Days 3, 7, and 14. No TEWL improvements during treatment phase were seen for the non-fragranced lotion.

TEWL for Lesion and Non-Lesion

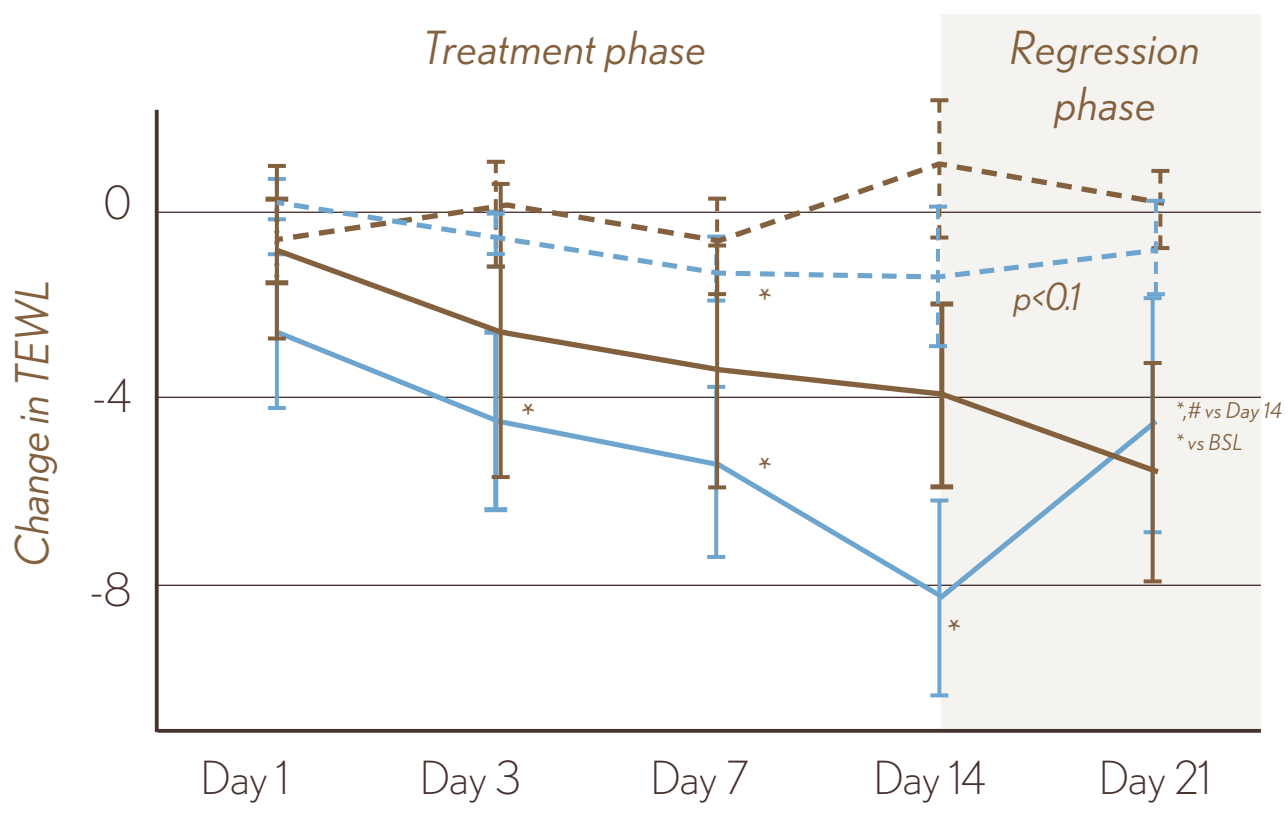
Treatment phase
Regression phase
Change in TEWL
0
-4
-8
p<0.1
*
*,# vs Day 14
* vs BSL
Day 1
Day 3
Day 7
Day 14
Day 21

- Treatment with AVEENO® Eczema Therapy Moisturizing Cream resulted in a statistically significant increase in skin hydration of lesional and non-lesional skin at Days 1, 3, 7, and 14. Treatment with AVEENO® Eczema Therapy Moisturizing Cream demonstrated a significantly greater improvement in skin hydration compared to the non-fragranced lotion (p<0.05).

Skin Hydration on Lesion and Non-Lesion Site

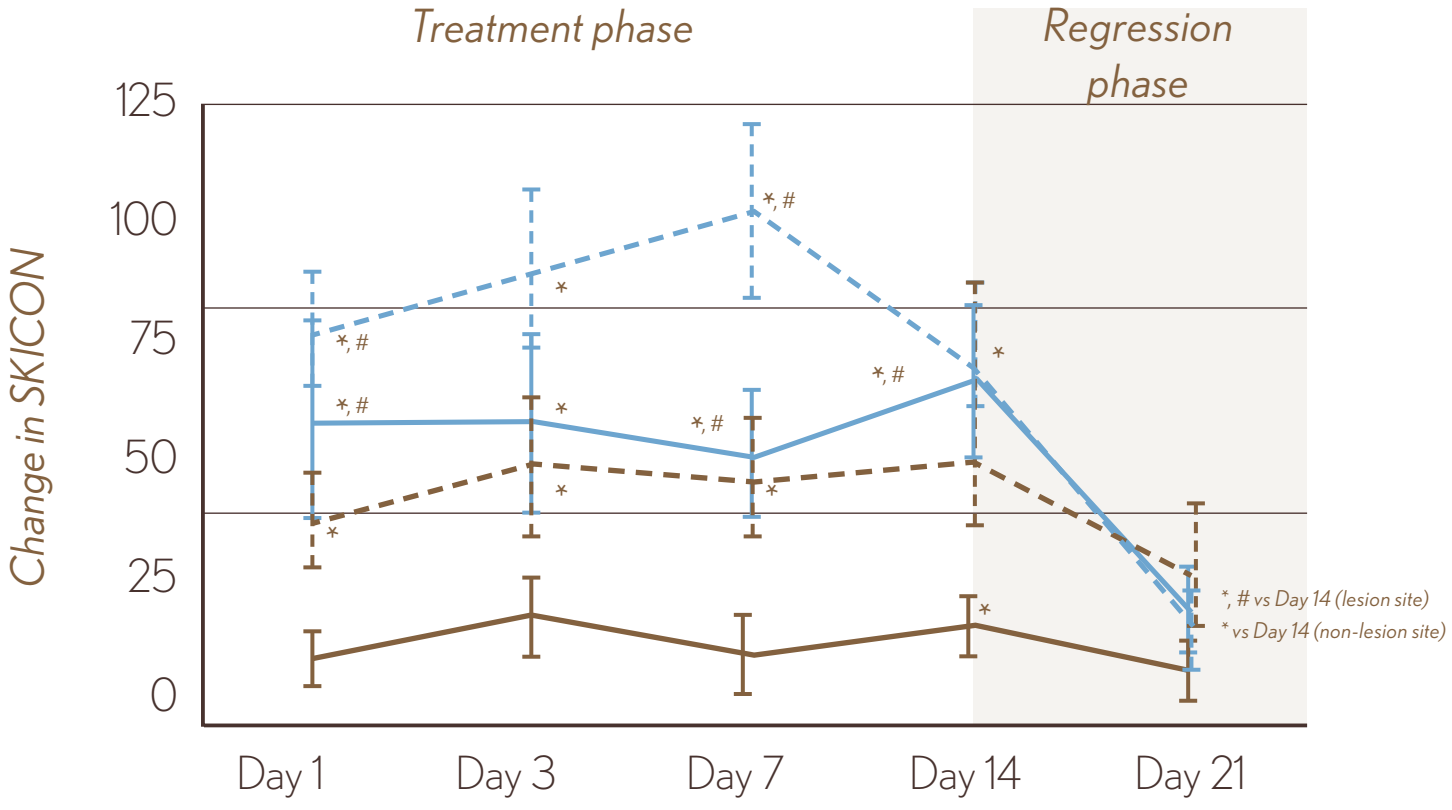
Treatment phase
Regression phase
Change in SKICON
125
100
75
50
25
0
*, #
*
*, # vs Day 14 (lesion site)
* vs Day 14 (non-lesion site)
Day 1
Day 3
Day 7
Day 14
Day 21

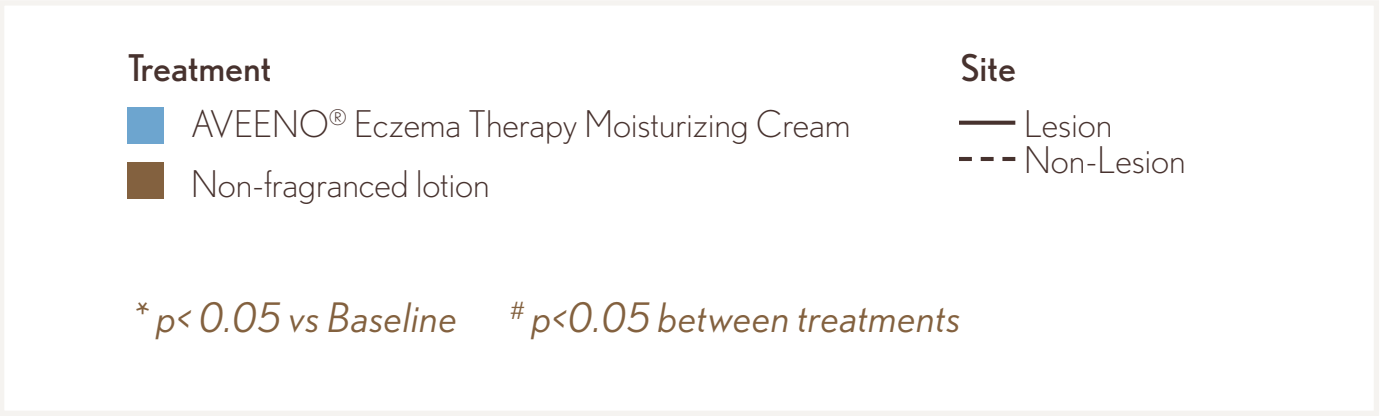
Treatment
AVEENO® Eczema Therapy Moisturizing Cream
Non-fragranced lotion
Site
Lesion
Non-Lesion
* p< 0.05 vs Baseline
p<0.05 between treatments

RESULTS (CONT)

FIGURE 2. ADSI and EASI Clinical Evaluation

- Treatment with AVEENO® Eczema Therapy Moisturizing Cream resulted in ~50% improvement in EASI clinical scoring at Day 14 (p<0.05).
- Treatment with AVEENO® Eczema Therapy Moisturizing Cream resulted in ~50% improvement in ADSI clinical scoring starting at Day 7 (p<0.05).
- Despite the withdrawal of treatment at Day 14, ADSI and EASI clinical scores remained improved from Baseline at Day 21, and were approximately equivalent to Day 3 and Day 7 values, respectively.

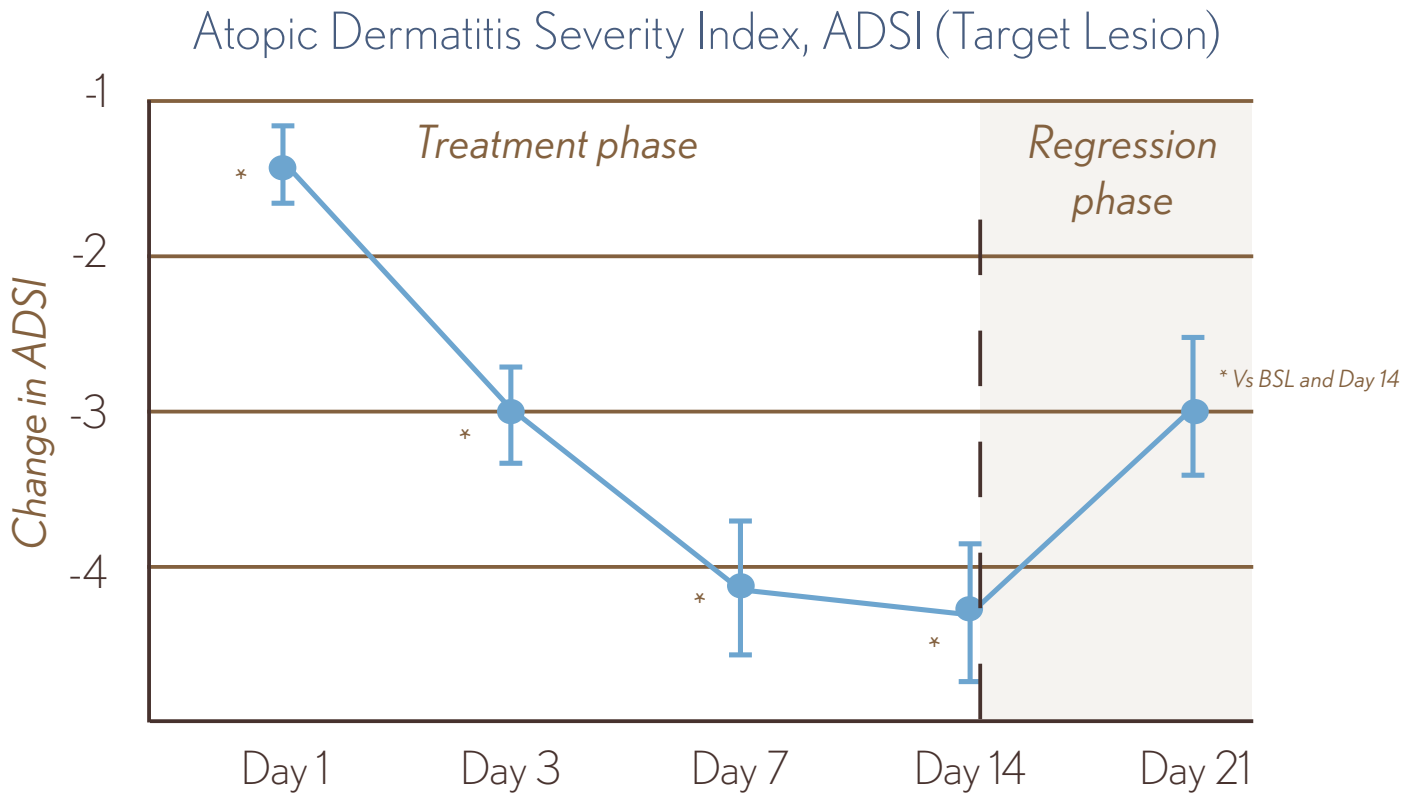

Atopic Dermatitis Severity Index, ADSI (Target Lesion)
Change in ADSI
-1
-2
-3
-4
Treatment phase
Regression phase
* Vs BSL and Day 14
Day 1
Day 3
Day 7
Day 14
Day 21

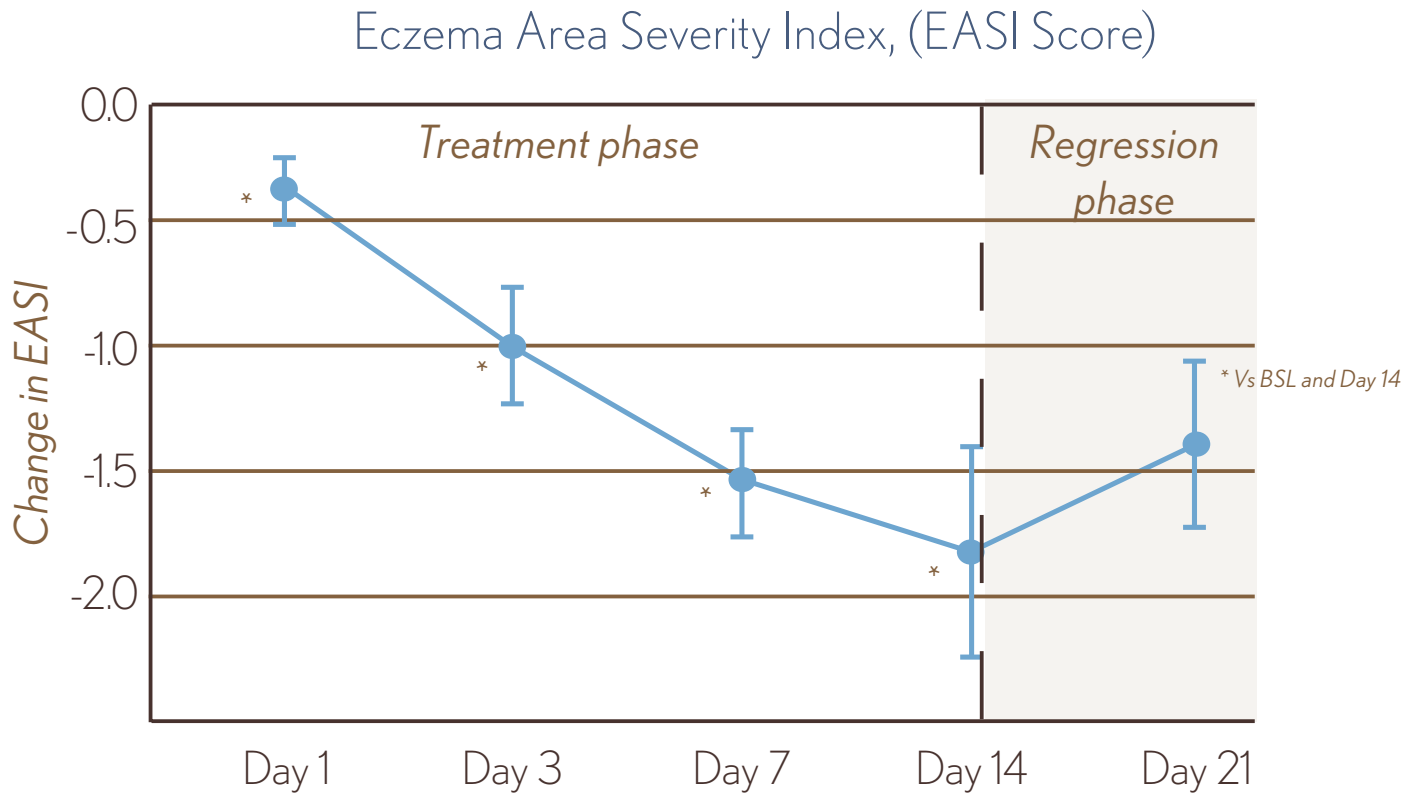

Eczema Area Severity Index, (EASI Score)
Change in EASI
0.0
-0.5
-1.0
-1.5
-2.0
Treatment phase
Regression phase
* Vs BSL and Day 14
Day 1
Day 3
Day 7
Day 14
Day 21

AVEENO® Eczema Therapy Moisturizing Cream * p <0.05 vs Baseline

FIGURE 3. VAS Measure of Itch

- Itch Severity improves starting at Day 1 (p<0.05) and continues throughout treatment, with a 60% improvement after 14 days.
- AVEENO® Eczema Therapy Moisturizing Cream provided significantly better itch reduction than the non-fragranced lotion (p<0.05) at Day 3.
- After 1 week withdrawal of both treatments, itch severity worsened vs. Day 14 but was still improved vs. Baseline (p<0.05).

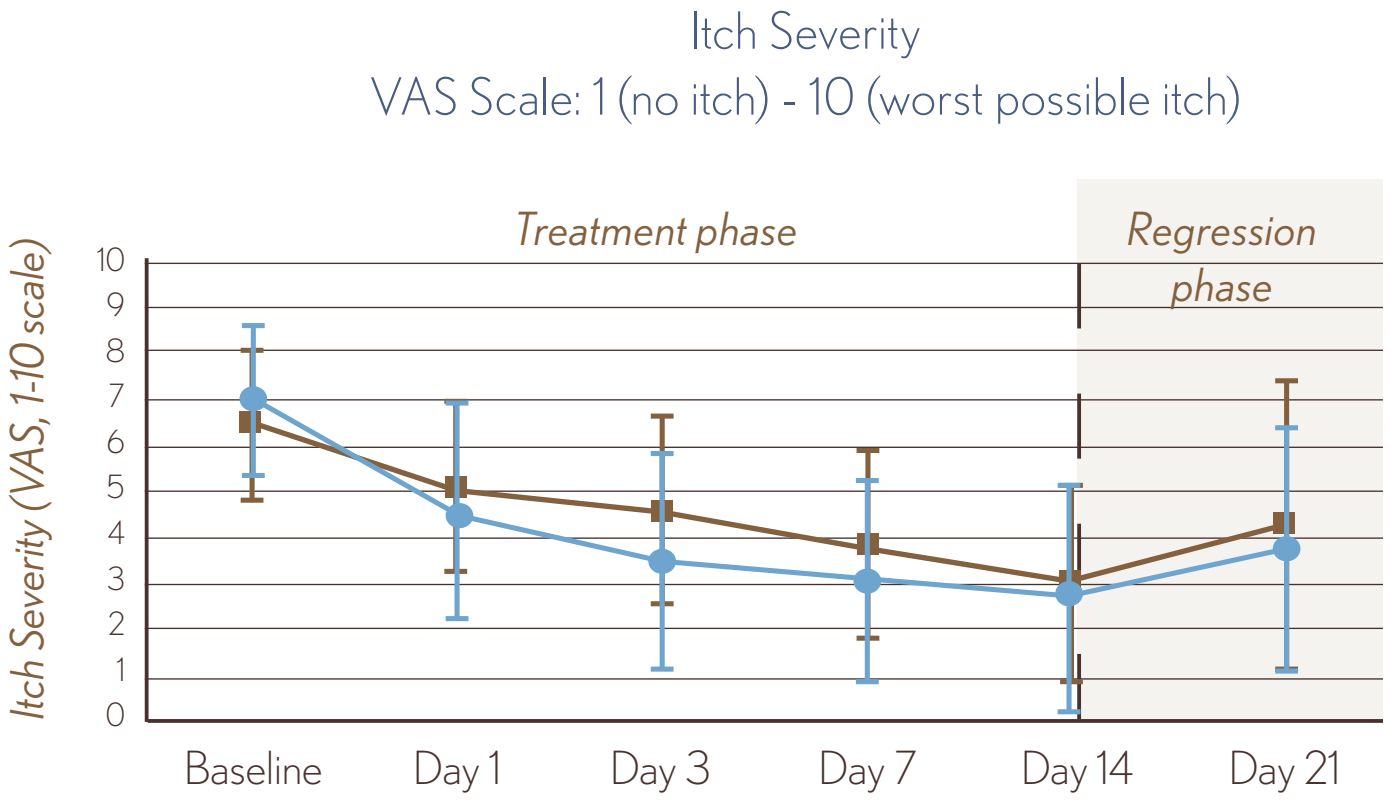

Itch Severity
VAS Scale: 1 (no itch) - 10 (worst possible itch)
Itch Severity (VAS, 1-10 scale)
10
9
8
7
6
5
4
3
2
1
0
Treatment phase
Regression phase
Baseline
Day 1
Day 3
Day 7
Day 14
Day 21

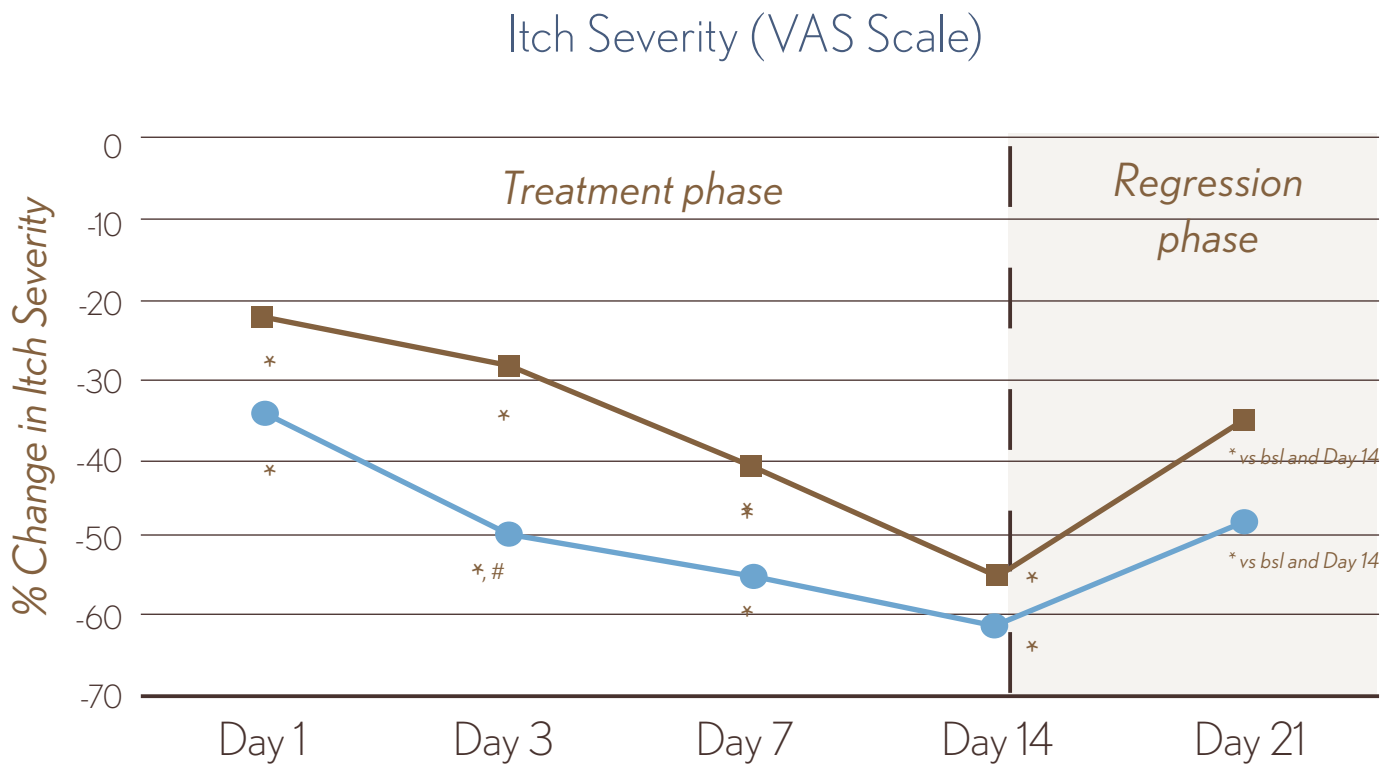

Itch Severity (VAS Scale)
% Change in Itch Severity
0
-10
-20
-30
-40
-50
-60
-70
Treatment phase
Regression phase
*, #
* vs bsl and Day 14
* vs bsl and Day 14
Day 1
Day 3
Day 7
Day 14
Day 21

AVEENO® Eczema Therapy Moisturizing Cream
Non-fragranced lotion

* p< 0.05 vs Baseline
p<0.05 between treatments

RESULTS (CONT)

FIGURE 4. Microbial Diversity at Baseline and Changes with Treatment

- At baseline, skin microbial communities of non-lesional skin had significantly greater diversity than lesional skin (P<0.05)
- For AVEENO® Eczema Therapy Moisturizing Cream, microbial diversity of lesional skin converged with non-lesional skin starting at Day 1, and remained similar throughout treatment & regression phases P>0.05.

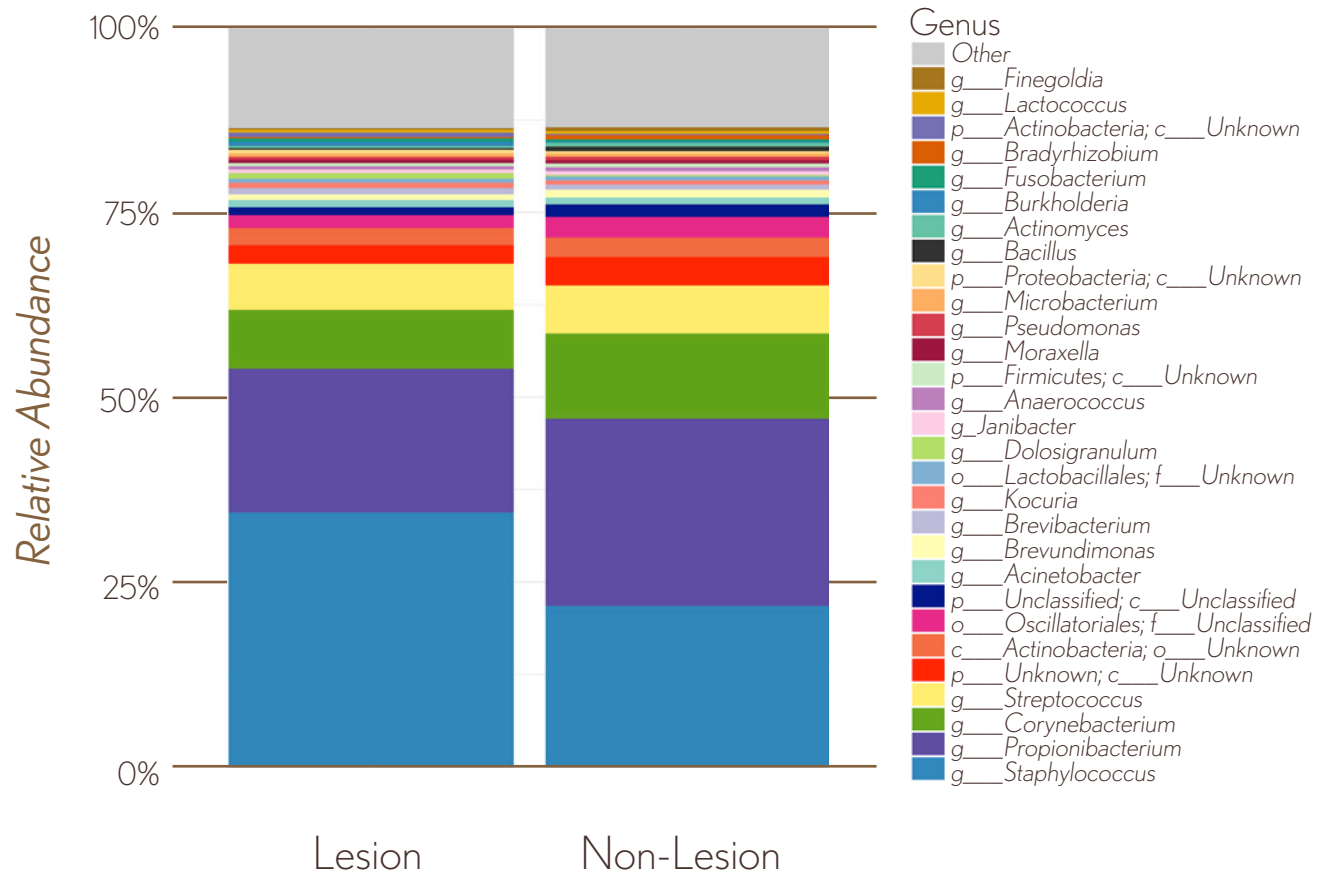

Relative Abundance
100%
75%
50%
25%
0%
Lesion
Non-Lesion
Genus
Other
g___Finegoldia
g___Lactococcus
p___Actinobacteria; c___Unknown
g___Bradyrhizobium
g___Fusobacterium
g___Burkholderia
g___Actinomyces
g___Bacillus
p___Proteobacteria; c___Unknown
g___Microbacterium
g___Pseudomonas
g___Moraxella
p___Firmicutes; c___Unknown
g___Anaerococcus
g_Janibacter
g___Dolosigranulum
o___Lactobacillales; f___Unknown
g___Kocuria
g___Brevibacterium
g___Brevundimonas
g___Acinetobacter
p___Unclassified; c___Unclassified
o___Oscillatoriales; f___Unclassified
c___Actinobacteria; o___Unknown
p___Unknown; c___Unknown
g___Streptococcus
g___Corynebacterium
g___Propionibacterium
g___Staphylococcus

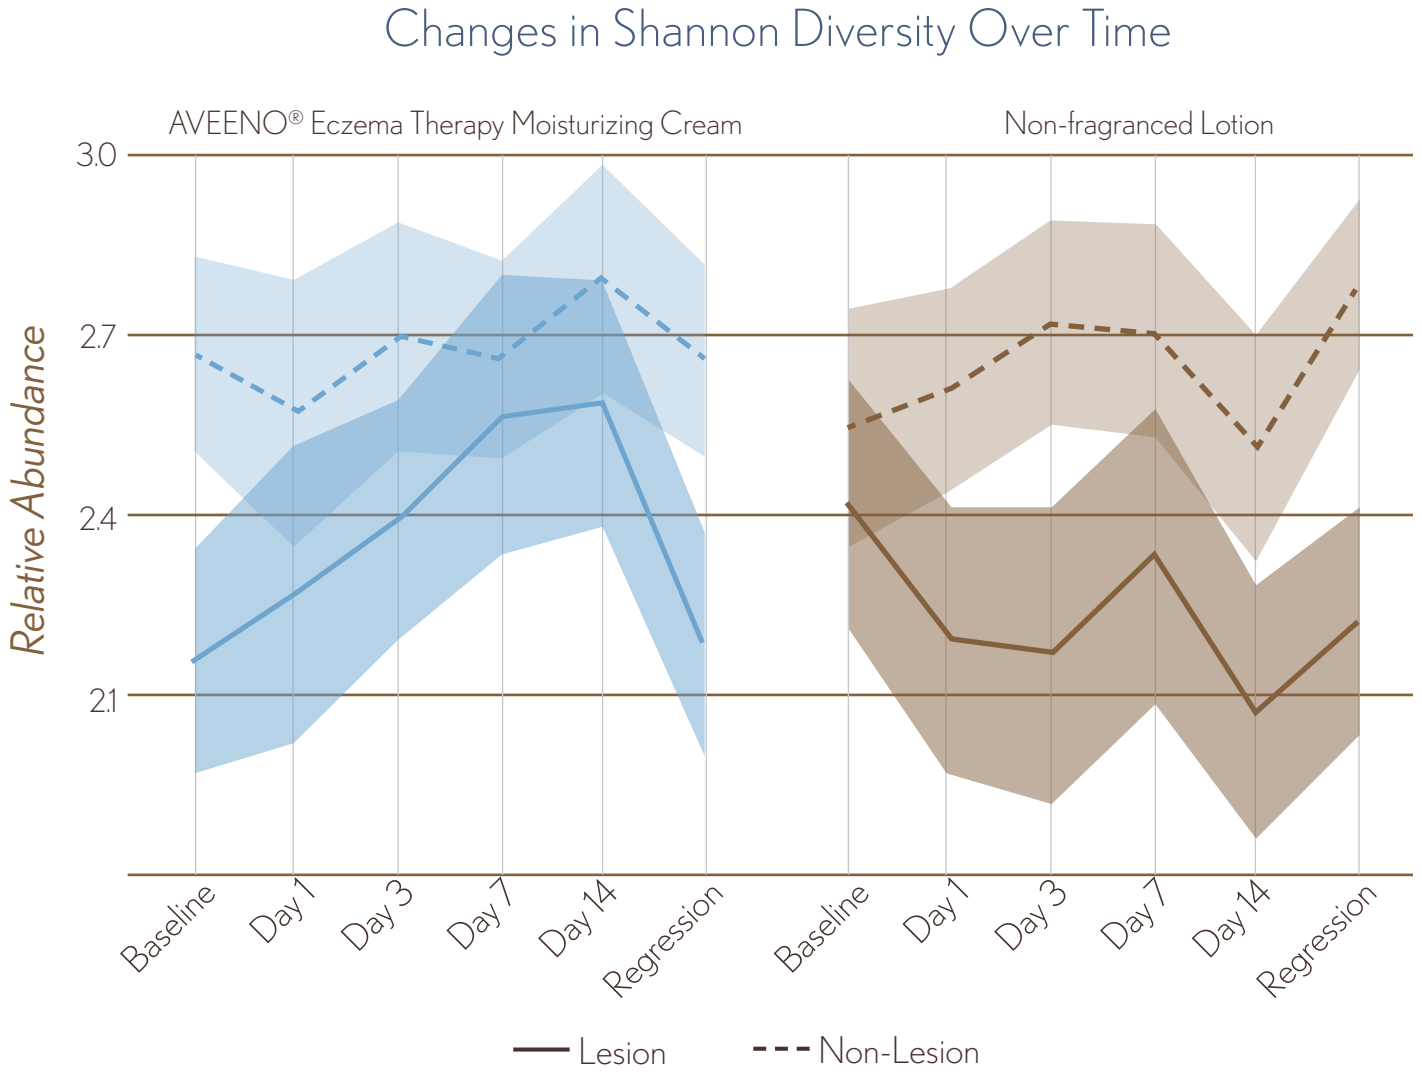

Changes in Shannon Diversity Over Time
AVEENO® Eczema Therapy Moisturizing Cream
Non-fragranced Lotion
Relative Abundance
3.0
2.7
2.4
2.1
Baseline
Day 1
Day 3
Day 7
Day 14
Regression
Baseline
Day 1
Day 3
Day 7
Day 14
Regression
Lesion
Non-Lesion

FIGURE 5. Average Differences in Lesional Microbial Diversity

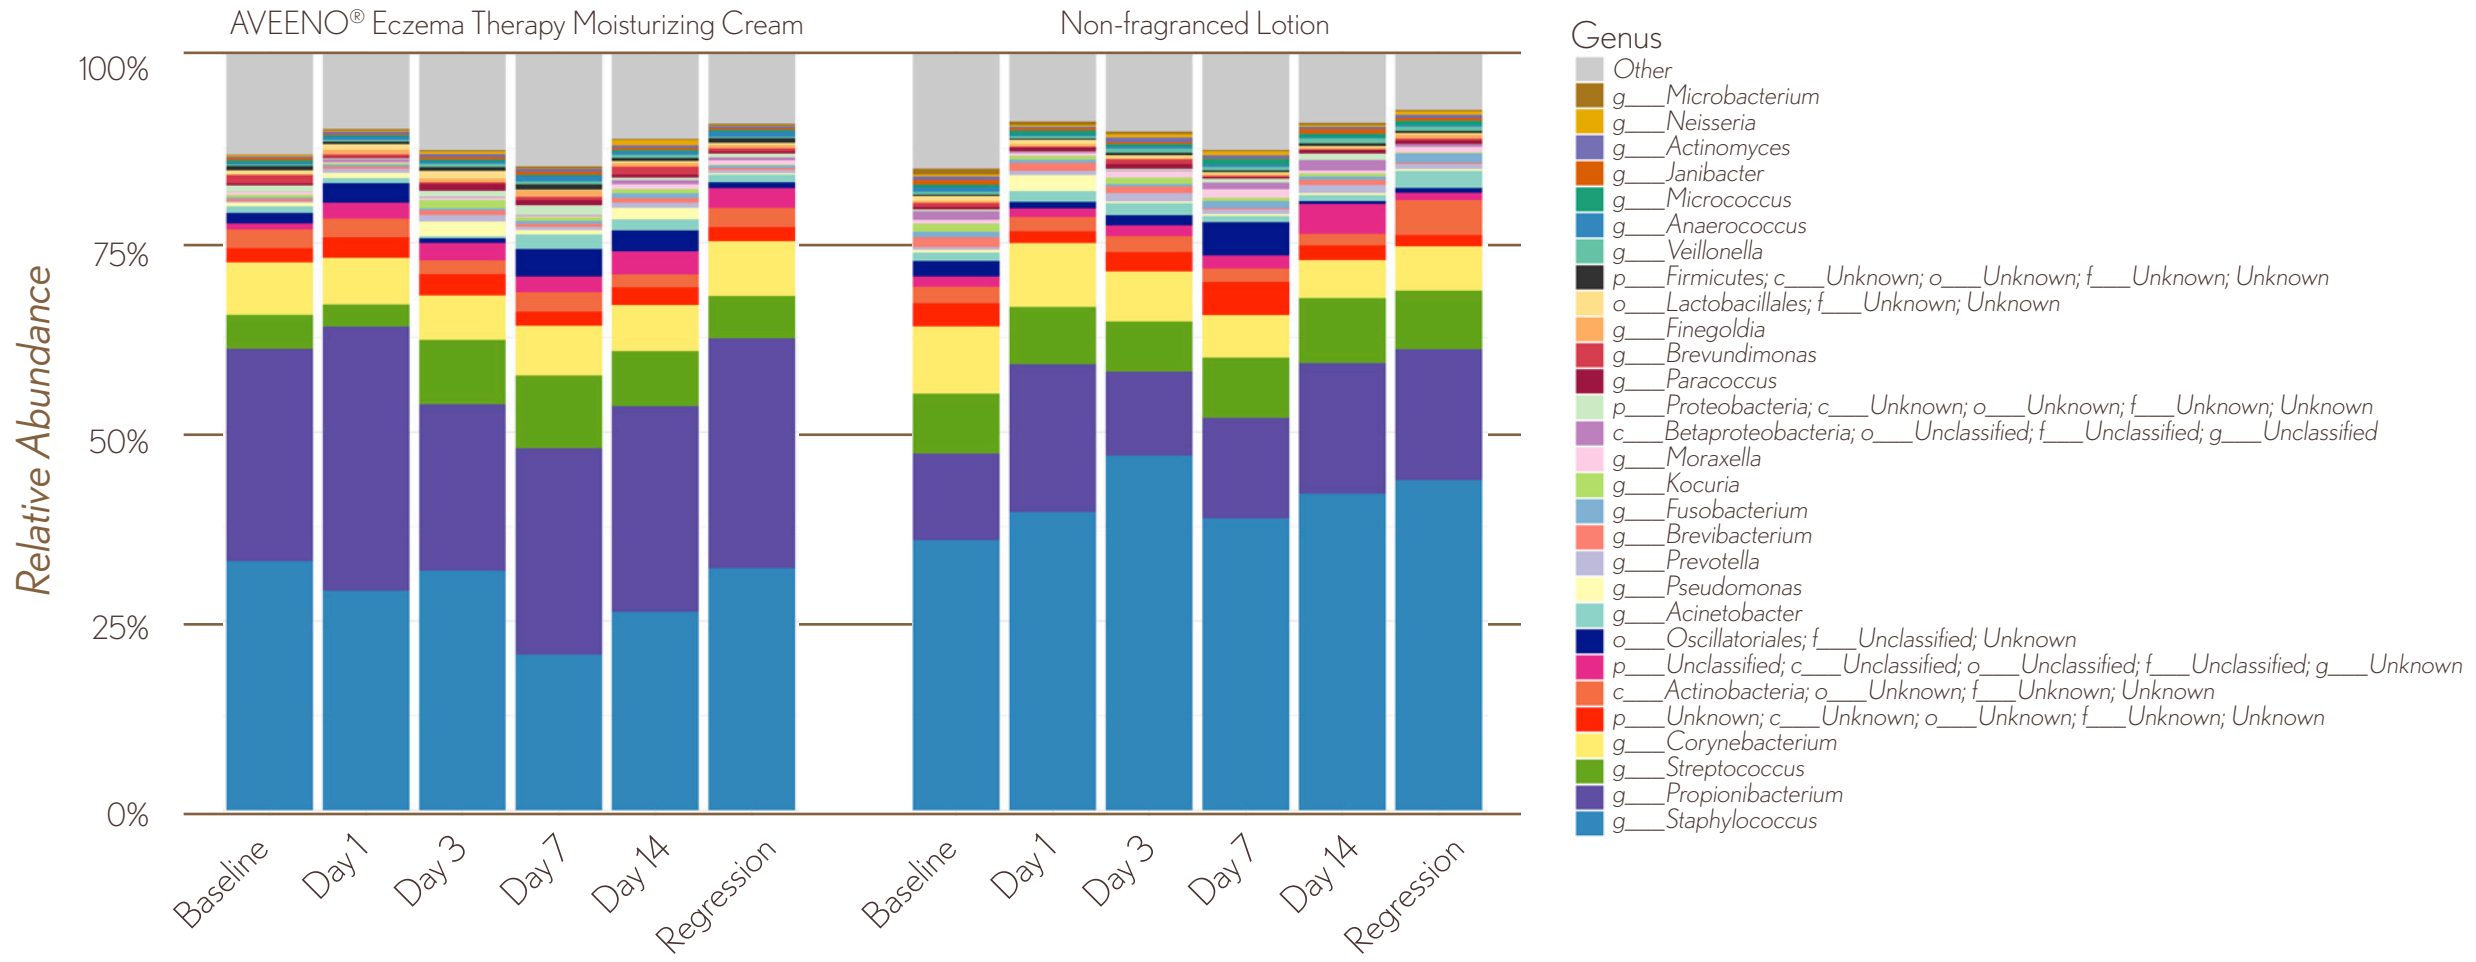

AVEENO® Eczema Therapy Moisturizing Cream
Non-fragranced Lotion
Relative Abundance
100%
75%
50%
25%
0%
Baseline
Day 1
Day 3
Day 7
Day 14
Regression
Baseline
Day 1
Day 3
Day 7
Day 14
Regression
Genus
Other
g___Microbacterium
g___Neisseria
g___Actinomyces
g___Janibacter
g___Micrococcus
g___Anaerococcus
g___Veillonella
p___Firmicutes; c___Unknown; o___Unknown; f___Unknown; Unknown
o___Lactobacillales; f___Unknown; Unknown
g___Finegoldia
g___Brevundimonas
g___Paracoccus
p___Proteobacteria; c___Unknown; o___Unknown; f___Unknown; Unknown
c___Betaproteobacteria; o___Unclassified; f___Unclassified; g___Unclassified
g___Moraxella
g___Kocuria
g___Fusobacterium
g___Brevibacterium
g___Prevotella
g___Pseudomonas
g___Acinetobacter
o___Oscillatoriales; f___Unclassified; Unknown
p___Unclassified; c___Unclassified; o___Unclassified; f___Unclassified; g___Unknown
c___Actinobacteria; o___Unknown; f___Unknown; Unknown
p___Unknown; c___Unknown; o___Unknown; f___Unknown; Unknown
g___Corynebacterium
g___Streptococcus
g___Propionibacterium
g___Staphylococcus

CONCLUSIONS

At baseline, lesional skin had a higher pH, TEWL and lower skin hydration than non-lesional skin. For subjects treated with AVEENO® Eczema Therapy Moisturizing Cream, disease severity significantly improved from baseline throughout the 14-day treatment. Both lesional and non-lesional skin had significant increases in hydration along with reduced pH, TEWL and itch, in contrast to minimal improvements following use of non-fragranced lotion. At baseline, skin microbial communities of non-lesional skin had significantly greater diversity than lesional skin. Throughout the 14-day treatment with AVEENO® Eczema Therapy Moisturizing Cream there were significant improvements in microbial diversity of lesional skin, with lesional skin microbial diversity reaching that of non-lesional skin by Day 1 and remaining similar throughout treatment and regression phases. These data confirm previous linkages between microbial diversity and AD severity, while extending current knowledge of various microbial populations present in AD lesional skin, which may aid in development of new microbiome-based therapies.

ACKNOWLEDGEMENTS

The authors would like to thank Jeremy Wilkinson at RTL Genomics, for skin microbiome sequencing and data analyses assistance.

© Johnson & Johnson Consumer Inc. 2017